MALDI Imaging Mass Spectrometry as a Tool to Evaluate Levels of ATP and its Metabolites in Mouse Tumor Models

Stephanie Dale[1], **Cristine Quiason-Huynh**[1], **Dewakar Sangaraju**[1], **Allan Jaochico**[1], **Meryssa Tran**[1], **Denise De Almeida Nagata**[2], **Yingyun Wang**[2], **Jill Shatner**[2], **Jane Grogan**[2]
[1]Department of Drug Metabolism & Pharmacokinetics, [2]Cancer Immunology, Genentech, Inc., 1 DNA Way, South San Francisco, CA 94080

INTRODUCTION

Cancer cells alter their metabolism to promote growth, survival, proliferation, and drug resistance. A common feature of this modified metabolism is the upregulation of glycolysis, the metabolism of glucose into energy in the form of ATP. In tumors and proliferating cells, the rate of glucose uptake and fermentation of glucose to lactate dramatically increases. Intratumoral extracellular ATP levels are elevated in a large range of tumors. Extracellular ATP is known to be unstable and can be hydrolyzed to its other metabolites including ADP and AMP. Furthermore, ATP and its metabolites have been studied to promote cancer cell growth, proliferation, survival rates, and modulate immune responses. Since ATP levels are unknown in pre-clinical tumor models, we developed a MALDI imaging mass spectrometry (IMS) assay to qualitatively visualize the levels of ATP and its metabolites in several tumor types including, CT26, E0771, EMT6, Hepa 1-6, and MC38. Quantitative LC-MS/MS experiments were also run in parallel to confirm the MALDI IMS results.

METHODS

Mouse tumors from 5 tumor models, CT26, E0771, EMT6, Hepa 1-6, and MC38 were harvested and placed in a 2mL Eppendorf micro tube, then immediately placed in liquid nitrogen. Tumors and spleens were sectioned at 12um thickness at the median level and thaw-mounted onto ITO-coated glass slides. Sections were spray-coated using the robotic HTX sprayer with 10 mg/mL 9-aminoacridine (9-AA) matrix in 70% ethanol with 4uM internal standard (~15 mL). Sections were analyzed by MALDI imaging on a 7.0T FTICR-MS in negative mode collecting 500 laser shots from m/z 100-650 at 100 um pixel resolution. Images were visualized in FlexImaging ver. 4.1 and normalized using root mean square. After MALDI IMS acquisition, matrix was rinsed off the tissue with 100% methanol and stained with hematoxylin and eosin.

RESULTS

LC-MS/MS Quantitation

Sample Name	ATP (ug/g)	Average ± S.D. (ug/g)	ADP (ug/g)	Average ± S.D. (ug/g)	AMP (ug/g)	Average ± S.D. (ug/g)
CT26 Tumor 1_1	2257		1371		1335	
CT26 Tumor 1_2	1683	1718 ± 522	1251	1103 ± 364	2195	1418 ± 739
CT26 Tumor 1_3	1215		688		723	
E0771 Tumor 2_1	420		534		3693	
E0771 Tumor 2-2	2903	1640 ±1242	1054	789 ± 260	2646	3167 ± 523
E0771 Tumor 2-3	1596		778		3188	
EMT6 Tumor 3_1	111		267		1697	
EMT6 Tumor 3_2	166	139 ± 28	497	390 ± 115	1097	1183 ± 476
EMT6 Tumor 3_3	140		406		756	
Hepa 1-6 Tumor 4_1	299		720		3906	
Hepa 1-6 Tumor 4_2	640	463 ± 170	1226	1055 ± 291	2850	3132 ± 679
Hepa 1-6 Tumor 4_3	450		1220		2640	
MC38 Tumor 5_1	232		340		2717	
MC38 Tumor 5_2	410	326 ± 89	596	518 ± 155	4471	3691 ± 893
MC38 Tumor 5_3	337		619		3885	

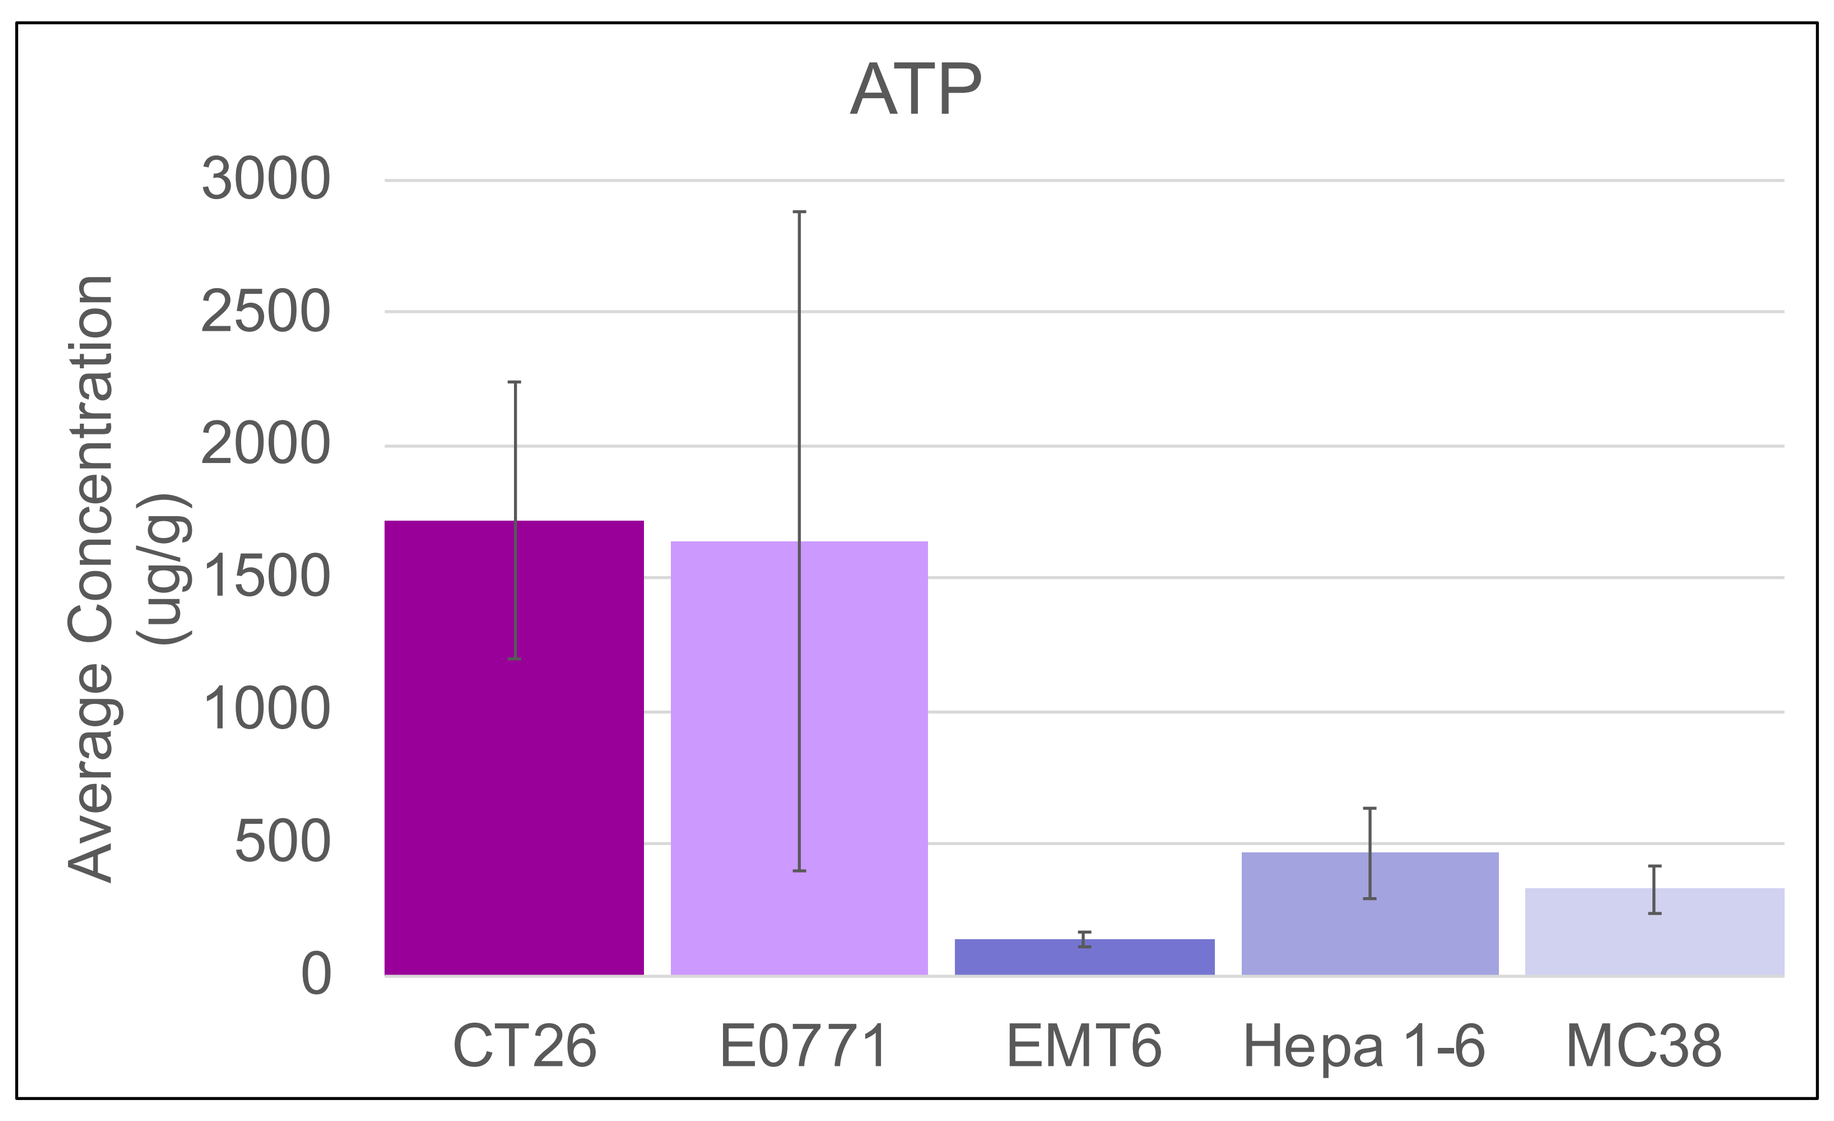
ATP
Average Concentration (ug/g)
3000
2500
2000
1500
1000
500
0
CT26
E0771
EMT6
Hepa 1-6
MC38

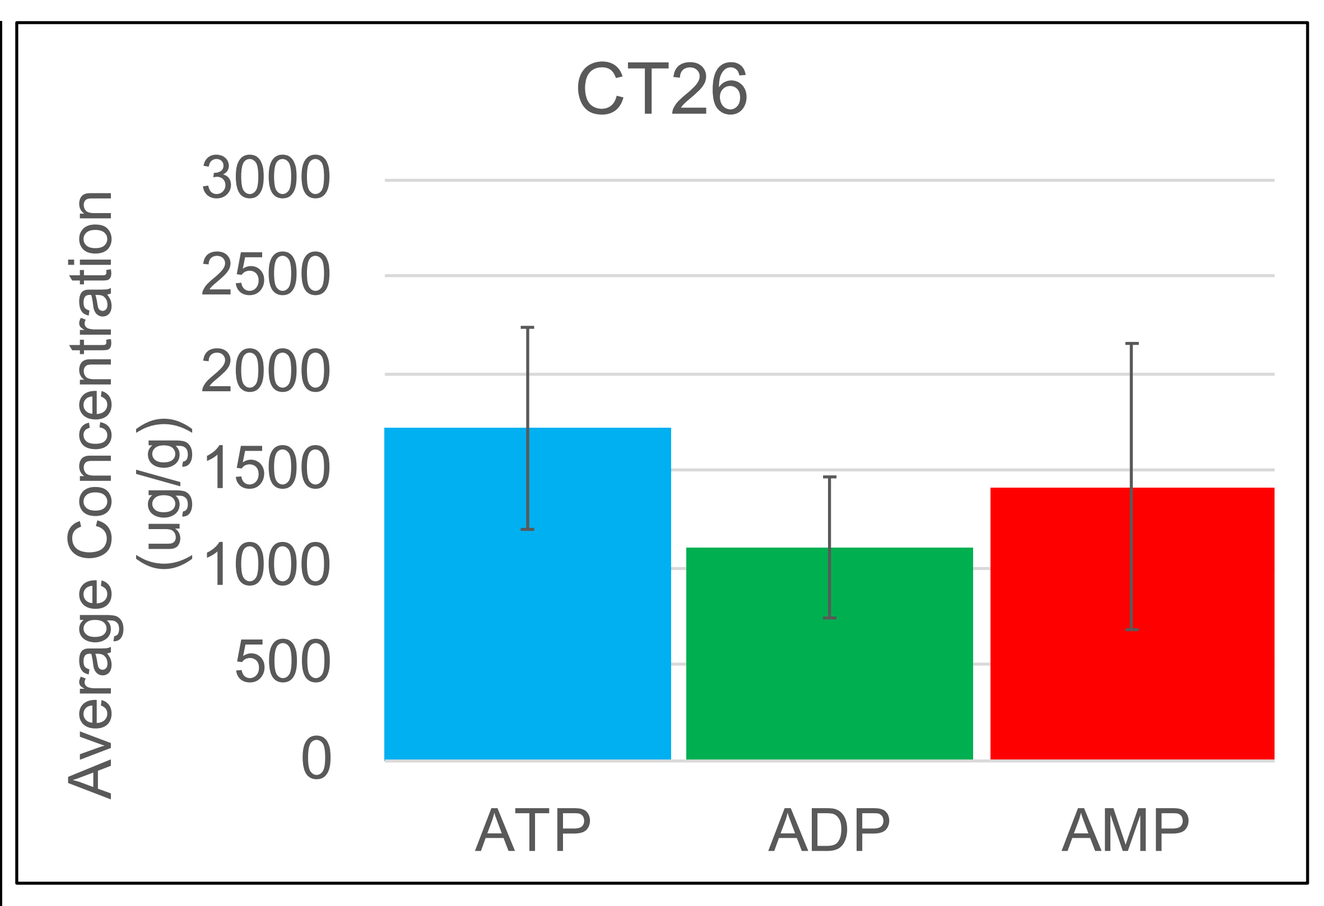
CT26
Average Concentration (ug/g)
3000
2500
2000
1500
1000
500
0
ATP
ADP
AMP

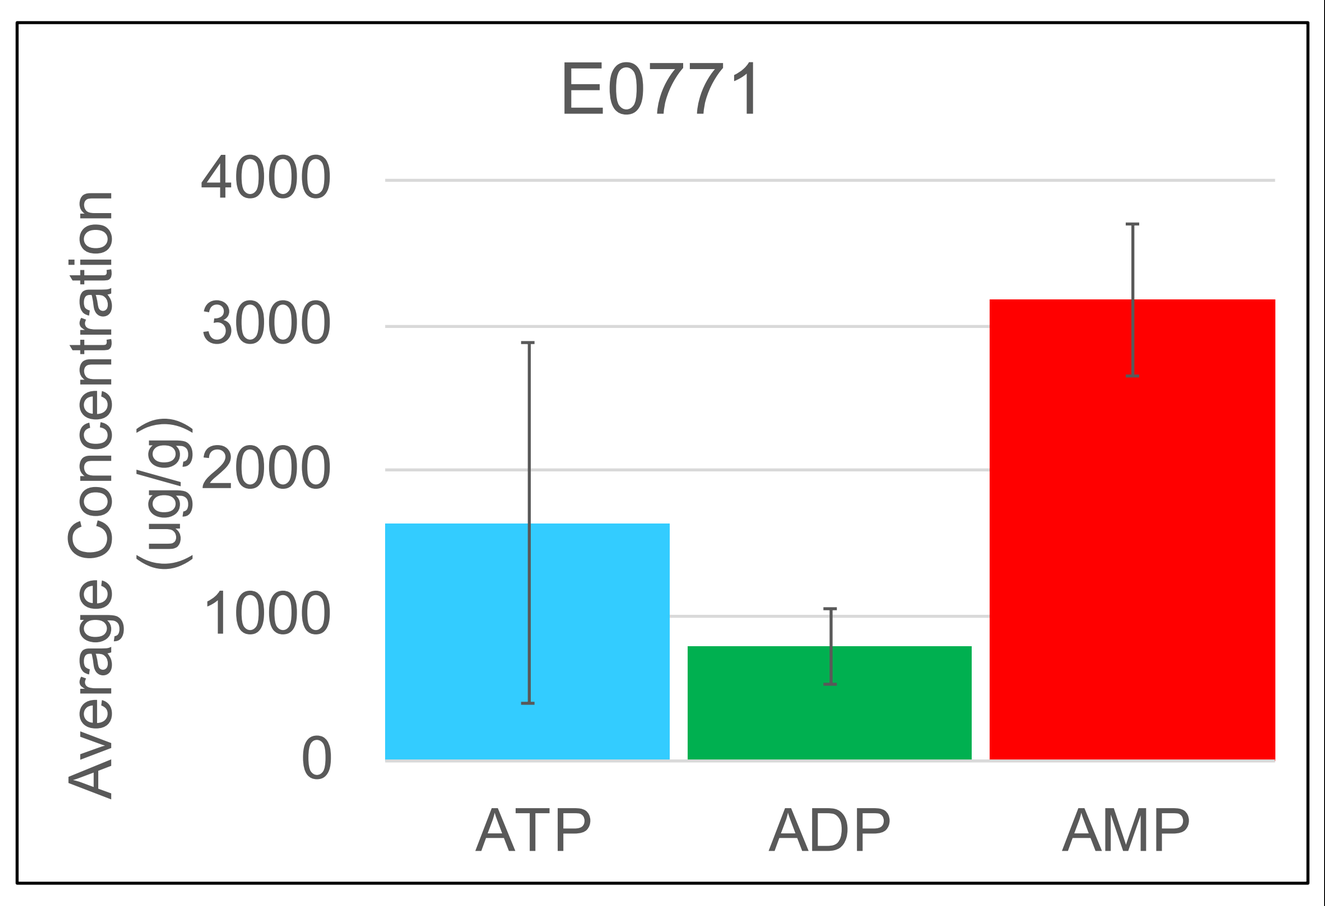
E0771
Average Concentration (ug/g)
4000
3000
2000
1000
0
ATP
ADP
AMP

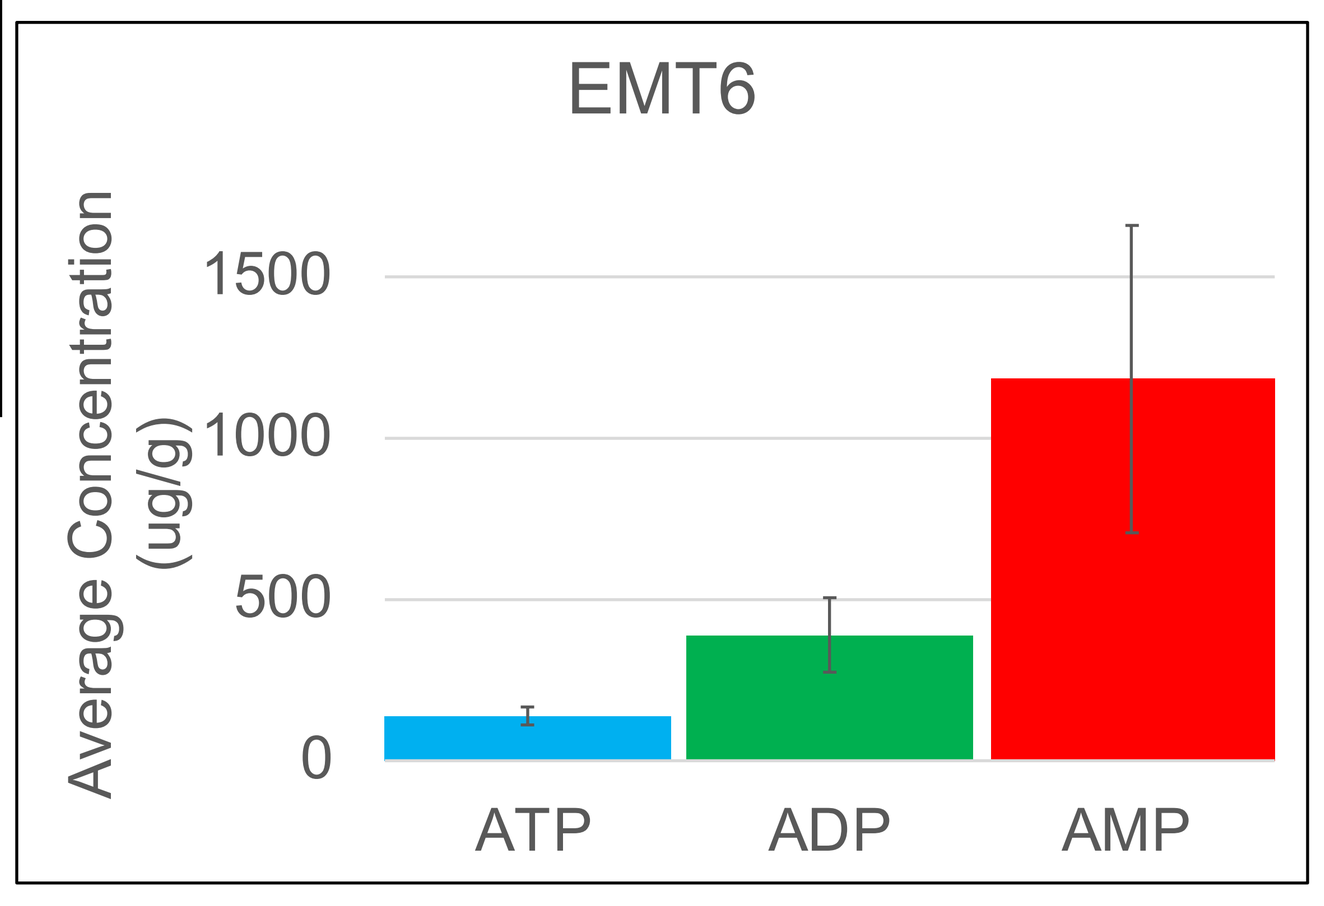
EMT6
Average Concentration (ug/g)
1500
1000
500
0
ATP
ADP
AMP

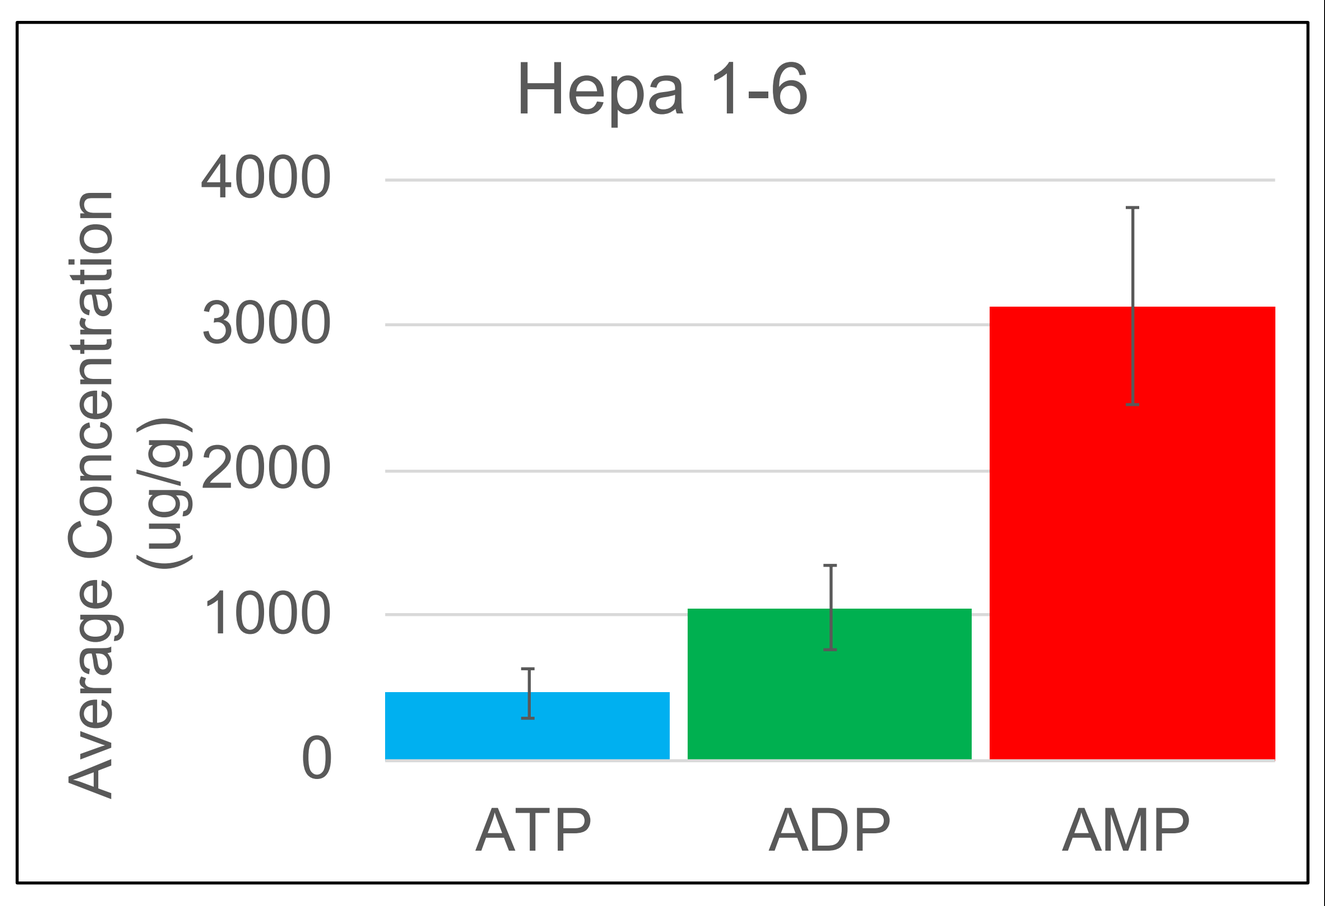
Hepa 1-6
Average Concentration (ug/g)
4000
3000
2000
1000
0
ATP
ADP
AMP

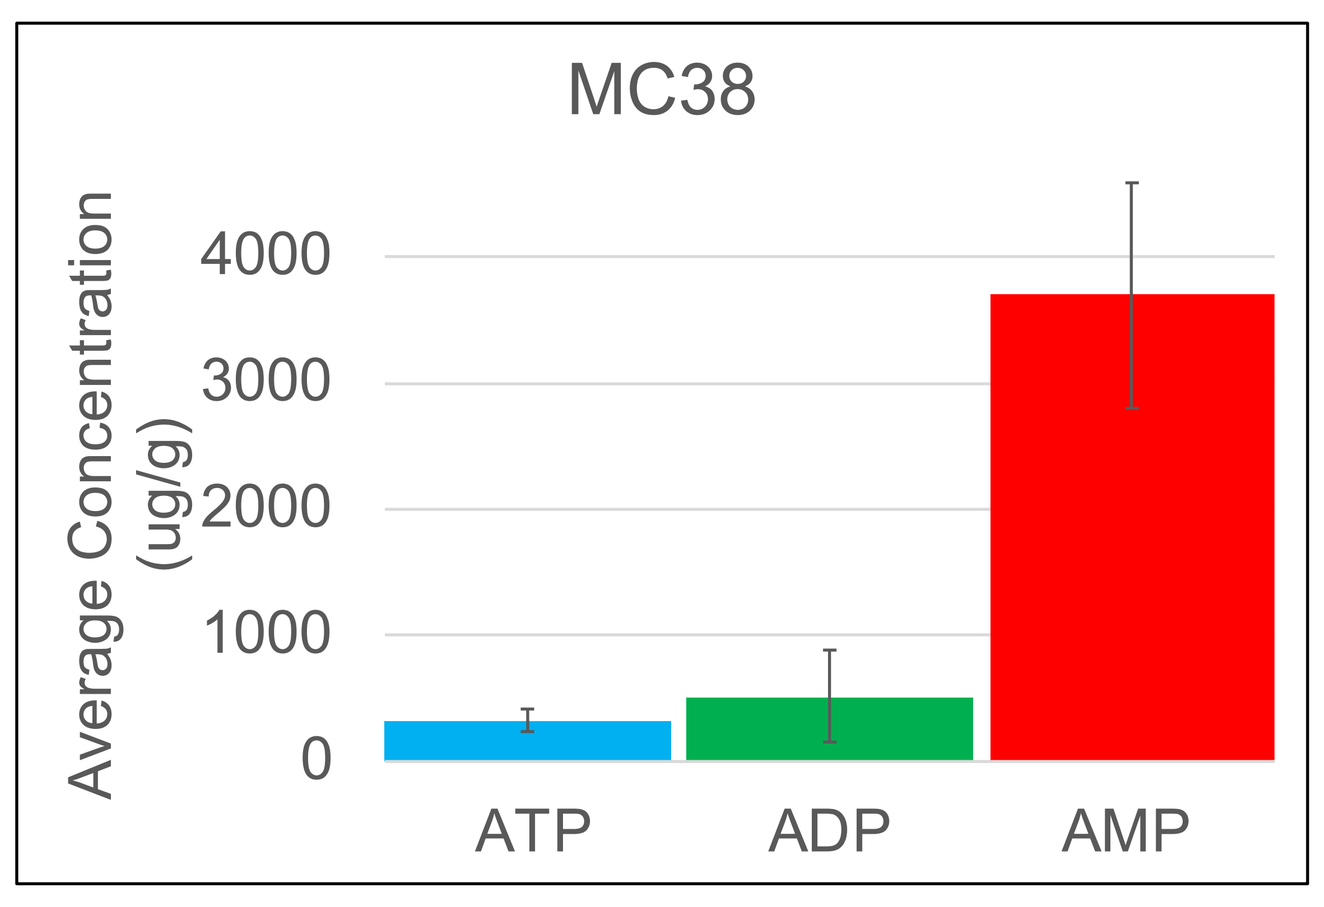
MC38
Average Concentration (ug/g)
4000
3000
2000
1000
0
ATP
ADP
AMP

Tumor Model | **H&E Stained Sections** | **MALDI MS Images**

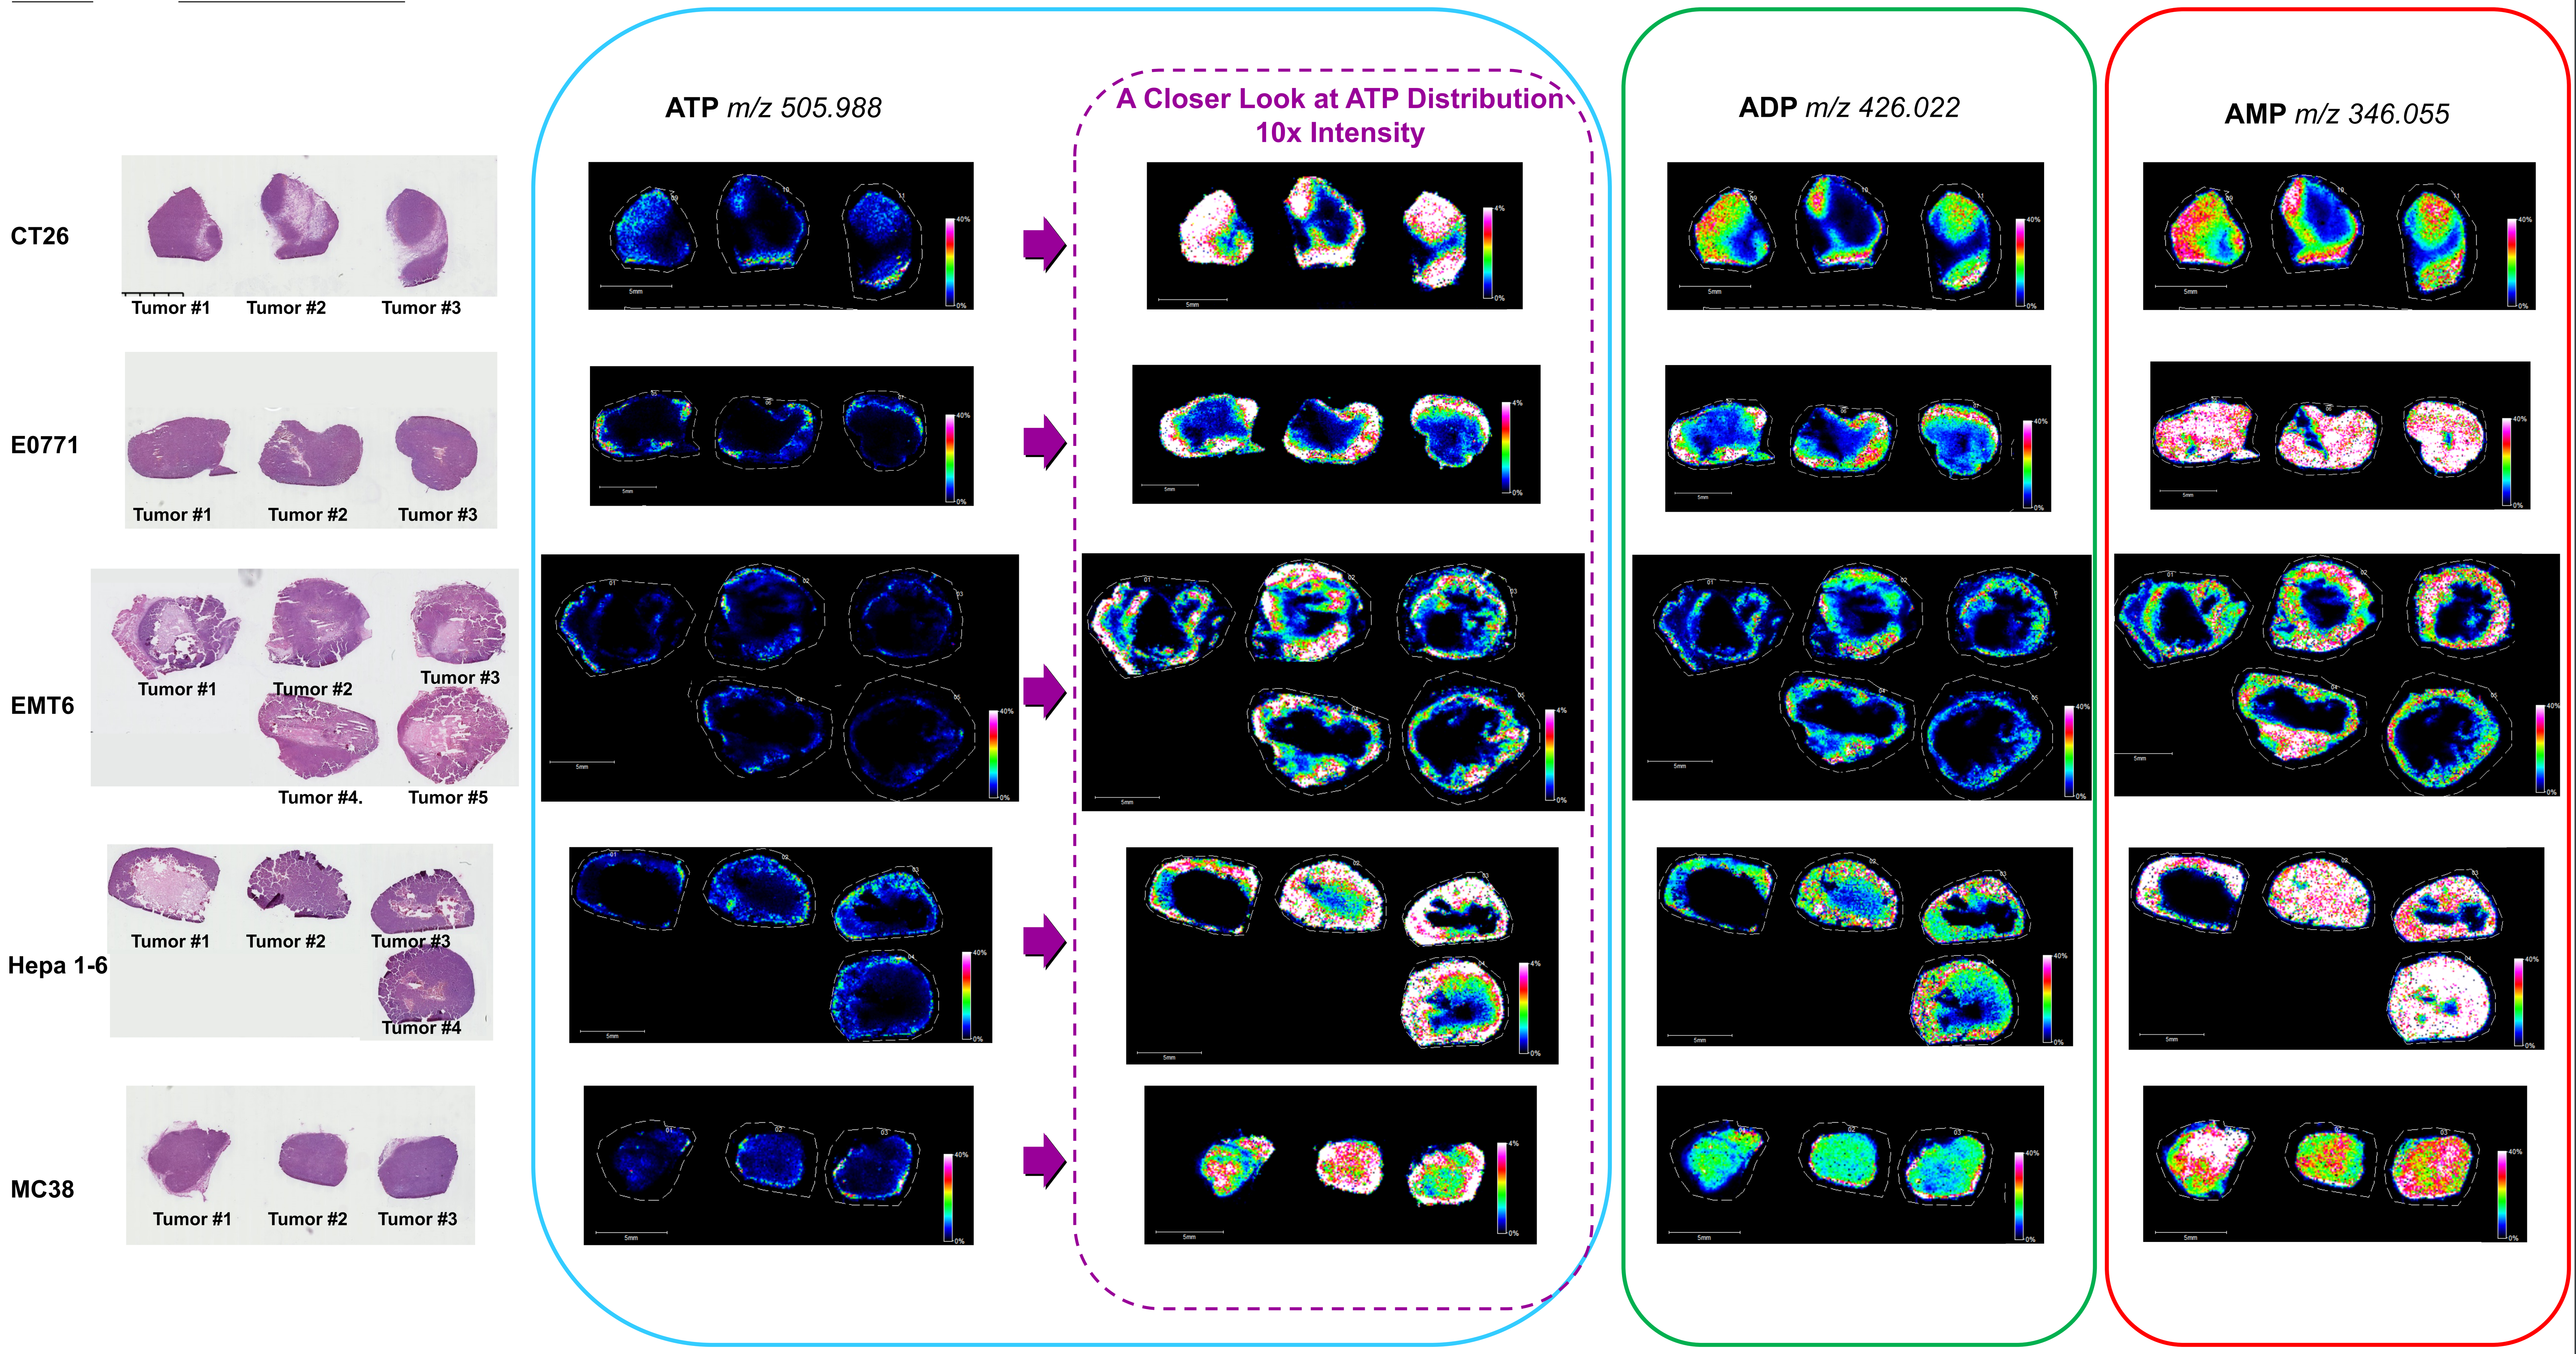
ATP m/z 505.988
A Closer Look at ATP Distribution
10x Intensity
ADP m/z 426.022
AMP m/z 346.055
CT26
Tumor #1
Tumor #2
Tumor #3
E0771
Tumor #1
Tumor #2
Tumor #3
EMT6
Tumor #1
Tumor #2
Tumor #3
Tumor #4.
Tumor #5
Hepa 1-6
Tumor #1
Tumor #2
Tumor #3
Tumor #4
MC38
Tumor #1
Tumor #2
Tumor #3

MALDI MS ATP Average Pixel Intensities

Tumor Model	Tumor #1	Tumor #2	Tumor #3	Tumor #4	Tumor #5	Average ± S.D.
CT26	0.13	0.07	0.11	-	-	0.1 ± 0.03
E0771	0.09	0.06	0.05	-	-	0.07 ± 0.02
EMT6	0.04	0.05	0.03	0.04	0.02	0.04 ± 0.01
Hepa 1-6	0.03	0.10	0.10	0.08	-	0.07 ± 0.03
MC38	0.05	0.11	0.10	-	-	0.09 ± 0.03

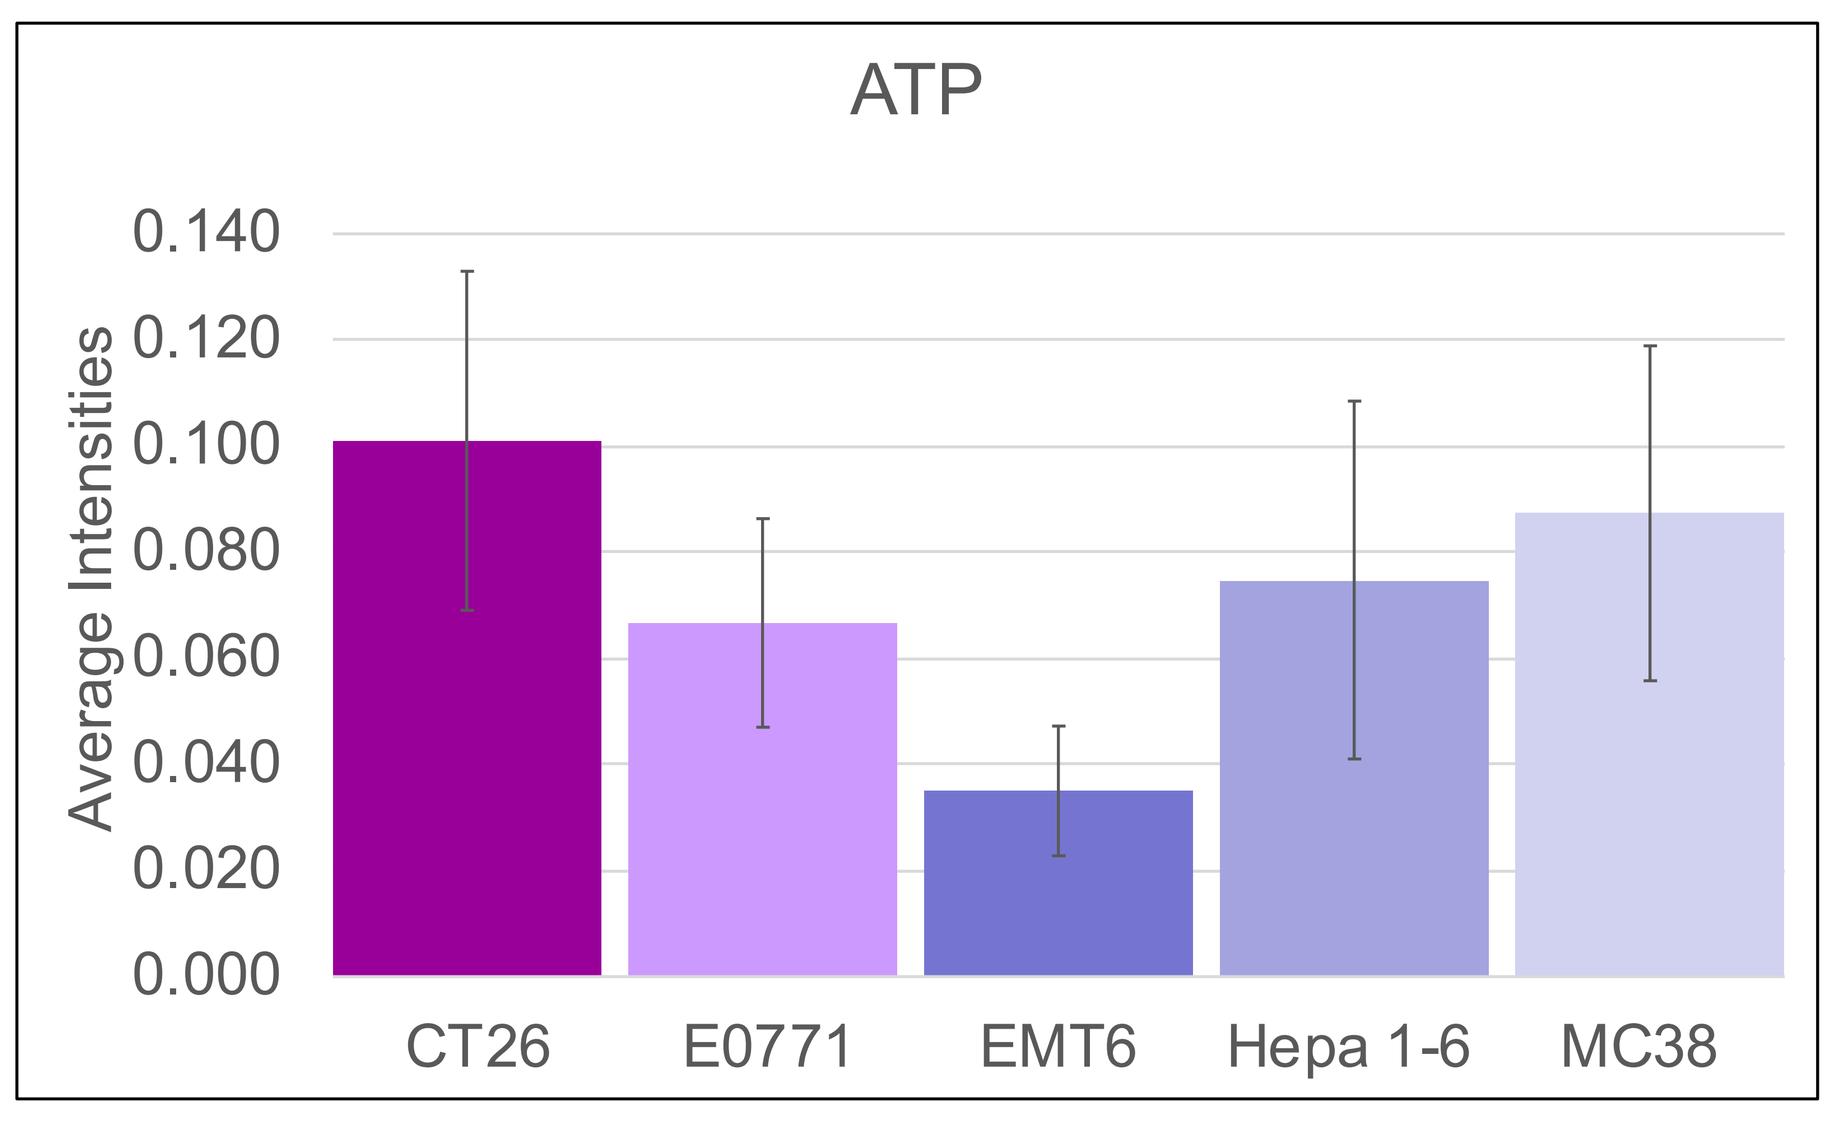
ATP
Average Intensities
0.140
0.120
0.100
0.080
0.060
0.040
0.020
0.000
CT26
E0771
EMT6
Hepa 1-6
MC38

CONCLUSIONS

Based on the LC-MS/MS quantitation data, the analyte concentration trends were ADP<AMP<ATP for CT26, ADP<ATP<AMP for E0771, and ATP<ADP<AMP for EMT6, Hepa 1-6, and MC38. By using MALDI IMS, we were able to detect ATP, ADP, and AMP in 5 pre-clinical tumor models. For the MALDI IMS results, the pixel intensities for each analyte were averaged in each tumor for every tumor model. Both technologies rank ordered tumor model CT26 with the highest level of ATP and EMT6 with the lowest ATP levels. The relative intensity levels in all tumor models were as follows: ATP<ADP<AMP. These findings correlate with LC-MS/MS quantitation of 3 tumor models, EMT6, Hepa 1-6, and MC38. Differences between the LC-MS/MS concentrations and MALDI MS average intensities for tumor models CT26 and E0771 could be due to several factors. It is possible that differences in tumor model biology, i.e. variations in lipid composition, can affect the ionization efficiency of the analytes of interest. It is also important to note that analyte distribution can vary throughout the entire tumor. For this experiment, tumors were only sectioned at the median level, which may not provide a complete representation of analyte distribution. For ATP, ADP, and AMP, distribution was heterogeneous in the tumors. Although LC-MS/MS experiments provided quantitative data, and matched the image results of 3 tumor models, it does not provide spatial information of the analytes of interest. These findings provide novel information of the distribution of ATP and its metabolites in CT26, E0771, EMT6, Hepa 1-6, and MC38 tumor models. This added layer of information, coupled with quantitation of ATP, ADP, and AMP from LC-MS/MS, can potentially further help elucidate disease pathways and biomarker discovery in the pre-clinical setting.

ACKOWLEDGEMENTS

Special thanks to the Metabolomics group in the DMPK Department and the Cancer Immunology Department at Genentech, Inc.